Renal Outcomes in People With HIV-1 and Renal Impairment Treated With B/F/TAF in Randomized Trials

859

Frank Post[1], David Wohl[2], Geoffroy Liegeon[3], Indira Brar[4], Debbie Hagins[5], Yazdan Yazdanpanah[6], Anchalee Avihingsanon[7], Hui Liu[8], Keith Aizen[8], Jason T Hindman[8], Samir Gupta[9]

[1]King's College Hospital NHS Foundation Trust, London, UK; [2]University of North Carolina, Chapel Hill, NC, USA; [3]Saint Louis-Hospital, AP-HP, Université Paris Cité, Paris, France; [4]Henry Ford Hospital, Detroit, MI, USA; [5]Chatham CARE Center, Savannah, GA, USA; [6]Bichat–Claude Bernard Hospital, AP-HP, Paris, France; [7]HIV-NAT, Thai Red Cross AIDS Research Centre, Bangkok, Thailand; [8]Gilead Sciences, Inc., Foster City, CA, USA; [9]Indiana University School of Medicine, Indianapolis, IN, USA

GS-US-380-1489/1490/1844/1878/1961/4030/4449/4458/4580

Conclusions

- In a retrospective analysis of renal outcomes in PWH with mild to moderate renal impairment ($eGFR_{CG}$ 30–< 90 mL/min) at baseline, $eGFR_{CG}$ remained stable with up to 5 years of B/F/TAF treatment
- Only 1 (< 0.1%) participant with renal impairment discontinued B/F/TAF due to a renal TEAE (acute kidney injury unrelated to B/F/TAF)
- Overall, renal TEAEs were infrequent, occurring in < 3% (28/1069) of participants with renal impairment; there were no occurrences of proximal renal tubulopathy or Fanconi syndrome
 - The rate of renal TEAEs was similar to that with other antiretroviral regimens[1-9]
- Changes in $eGFR_{CG}$ and the occurrence of renal safety events were generally consistent between those with renal impairment at baseline versus those without renal impairment ($eGFR_{CG}$ ≥ 90 mL/min)

Plain Language Summary

- Many people living with the human immunodeficiency virus (HIV) have conditions that affect their kidneys
- This analysis combined information from nine different studies. It looked at whether the HIV treatment bictegravir/emtricitabine/tenofovir alafenamide (B/F/TAF) is safe for people with HIV and kidney disease
- This study looked at over 1000 people who had mildly to moderately reduced kidney function and were taking B/F/TAF. The researchers examined how kidney function changed over time while taking this medication
 - The study also looked to see if anyone developed a new kidney-related condition and if anyone had to stop taking B/F/TAF because of these conditions
- The results showed that kidney function remained the same and did not worsen with B/F/TAF use in people with HIV who have conditions affecting their kidneys
 - Kidney function after taking B/F/TAF was the same as in people with HIV taking B/T/TAF who did not have conditions affecting their kidneys
- Only one person stopped taking B/F/TAF because of a new condition affecting their kidneys; the doctors did not think this condition was caused by B/F/TAF
- Less than 3% of people in this study had a new condition related to their kidneys while taking B/F/TAF
- This suggests B/F/TAF could be a good option for people with HIV whose kidney function was mildly to moderately reduced

Introduction

- People with HIV (PWH) tend to experience more comorbidities with age than people without HIV[10]
- Chronic kidney disease (CKD) is a common comorbidity in PWH,[11] and its prevalence among PWH is likely to increase as the population gets older
- With the need for long-term antiretroviral therapy in PWH, it is critical to establish the renal safety of HIV treatments
- Tenofovir alafenamide–based regimens have a superior renal safety profile compared with tenofovir disoproxil fumarate–based regimens,[12,13] improving treatment options for PWH with mild to moderate renal impairment[14]

Objective

- To assess renal outcomes through 5 years in PWH with mild to moderate renal impairment receiving bictegravir/emtricitabine/tenofovir alafenamide (B/F/TAF), using pooled data from nine studies

Methods

Study Design

B/F/TAF Phase 3/3b randomized trials[a]

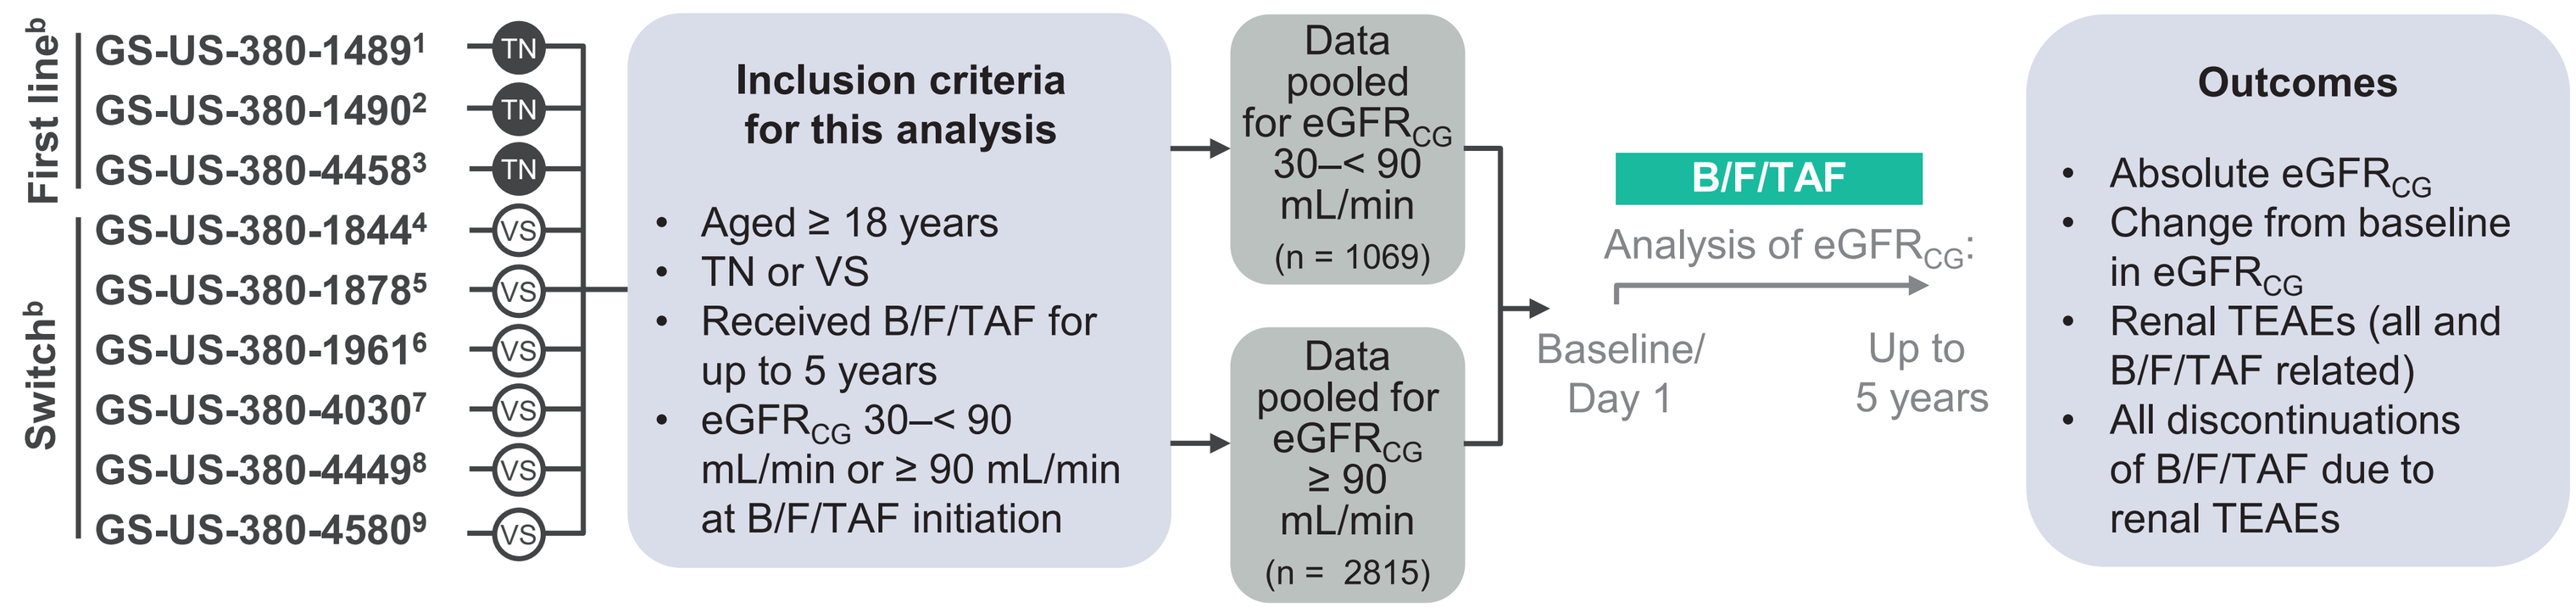
First line[b]
GS-US-380-1489[1] TN
GS-US-380-1490[2] TN
GS-US-380-4458[3] TN
Switch[b]
GS-US-380-1844[4] VS
GS-US-380-1878[5] VS
GS-US-380-1961[6] VS
GS-US-380-4030[7] VS
GS-US-380-4449[8] VS
GS-US-380-4580[9] VS
Inclusion criteria for this analysis
• Aged ≥ 18 years
• TN or VS
• Received B/F/TAF for up to 5 years
• eGFRCG 30–< 90 mL/min or ≥ 90 mL/min at B/F/TAF initiation
Data pooled for eGFRCG 30–< 90 mL/min (n = 1069)
Data pooled for eGFRCG ≥ 90 mL/min (n = 2815)
B/F/TAF
Analysis of eGFRCG:
Baseline/ Day 1
Up to 5 years
Outcomes
• Absolute eGFRCG
• Change from baseline in eGFRCG
• Renal TEAEs (all and B/F/TAF related)
• All discontinuations of B/F/TAF due to renal TEAEs

[a]GS-US-380-4449 was not randomized (single-group study). [b]Includes participants randomized to comparator group during the randomized phase who switched to B/F/TAF in the extension phase. B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; $eGFR_{CG}$, estimated glomerular filtration rate by Cockcroft-Gault equation; TEAE, treatment-emergent adverse event; TN, treatment-naïve; VS, virologically suppressed.

- Data from nine B/F/TAF Phase 3/3b trials were pooled:
 - Eight randomized studies: NCT02607930 (GS-US-380-1489),[1] NCT02607956 (GS-US-380-1490),[2] NCT03547908 (GS-US-380-4458),[3] NCT02603120 (GS-US-380-1844),[4] NCT02603107 (GS-US-380-1878),[5] NCT02652624 (GS-US-380-1961),[6] NCT03110380 (GS-US-380-4030),[7] NCT03631732 (GS-US-380-4580)[9]
 - One single-group study: NCT03405935 (GS-US-380-4449)[8]

Results

Baseline Demographics and Disease Characteristics by Baseline $eGFR_{CG}$

	30–< 90 mL/min (Renal Impairment)				≥ 90 mL/min (No Renal Impairment) n = 2815
	30–< 45 mL/min (Stage 3b $eGFR_{CG}$) n = 8	45–< 60 mL/min (Stage 3a $eGFR_{CG}$) n = 86	60–< 90 mL/min (Stage 2 $eGFR_{CG}$) n = 975	Overall n = 1069	
Age, years, median (Q1, Q3)	70 (61, 75)	62 (54, 67)	52 (43, 59)	53 (44, 60)	39 (31, 49)
Female sex at birth, n (%)	2 (25)	34 (40)	296 (30)	332 (31)	642 (23)
Race, n (%)					
White	5 (63)	45 (52)	492 (51)	542 (51)	1374 (49)
Black	2 (25)	33 (38)	321 (33)	356 (33)	998 (35)
Asian	0	5 (6)	110 (11)	115 (11)	240 (9)
Other[a]	1 (13)	3 (3)	52 (5)	56 (5)	203 (7)
Hispanic or Latine ethnicity, n (%)	1 (13)	12 (14)	148 (15)	161 (15)	529 (19)
$eGFR_{CG}$, mL/min, median (Q1, Q3)	40.8 (38.6, 42.5)	55.8 (51.0, 58.2)	78.6 (72.0, 84.8)	77.7 (69.6, 84.6)	116.8 (102.7, 136.8)
Treatment history, n (%)					
Treatment naïve	0	5 (6)	79 (8)	84 (8)	670 (24)
Virologically suppressed	8 (100)	81 (94)	896 (92)	985 (92)	2145 (76)

[a]Includes American Indian or Alaska Native; Native Hawaiian or Other Pacific Islander; other race; and participants with Not Permitted designation (when local regulators did not allow collection of race/ethnicity information). $eGFR_{CG}$, estimated glomerular filtration rate by Cockcroft-Gault equation; Q, quartile.

Baseline Characteristics

- Participants with more advanced CKD had a higher median age than those with less advanced CKD or no renal impairment

Treatment Duration With B/F/TAF

- Median (quartile [Q]1, Q3) duration of B/F/TAF treatment was 83.4 (52.4, 101.9) weeks for the overall study population with renal impairment (Stage 2-3b $eGFR_{CG}$) and 96.0 (64.1, 131.9) weeks for those without renal impairment
- By $eGFR_{CG}$ stage, median (Q1, Q3) [range] treatment duration was:
 - Stage 2 $eGFR_{CG}$: 84.3 (52.4, 103.4) [0.1-277.0] weeks
 - Stage 3a $eGFR_{CG}$: 72.5 (52.1, 96.0) [2.1-259.3] weeks
 - Stage 3b $eGFR_{CG}$: 95.4 (70.3, 101.8) [60.0-111.3] weeks

$eGFR_{CG}$ Change From Baseline Through 5 Years

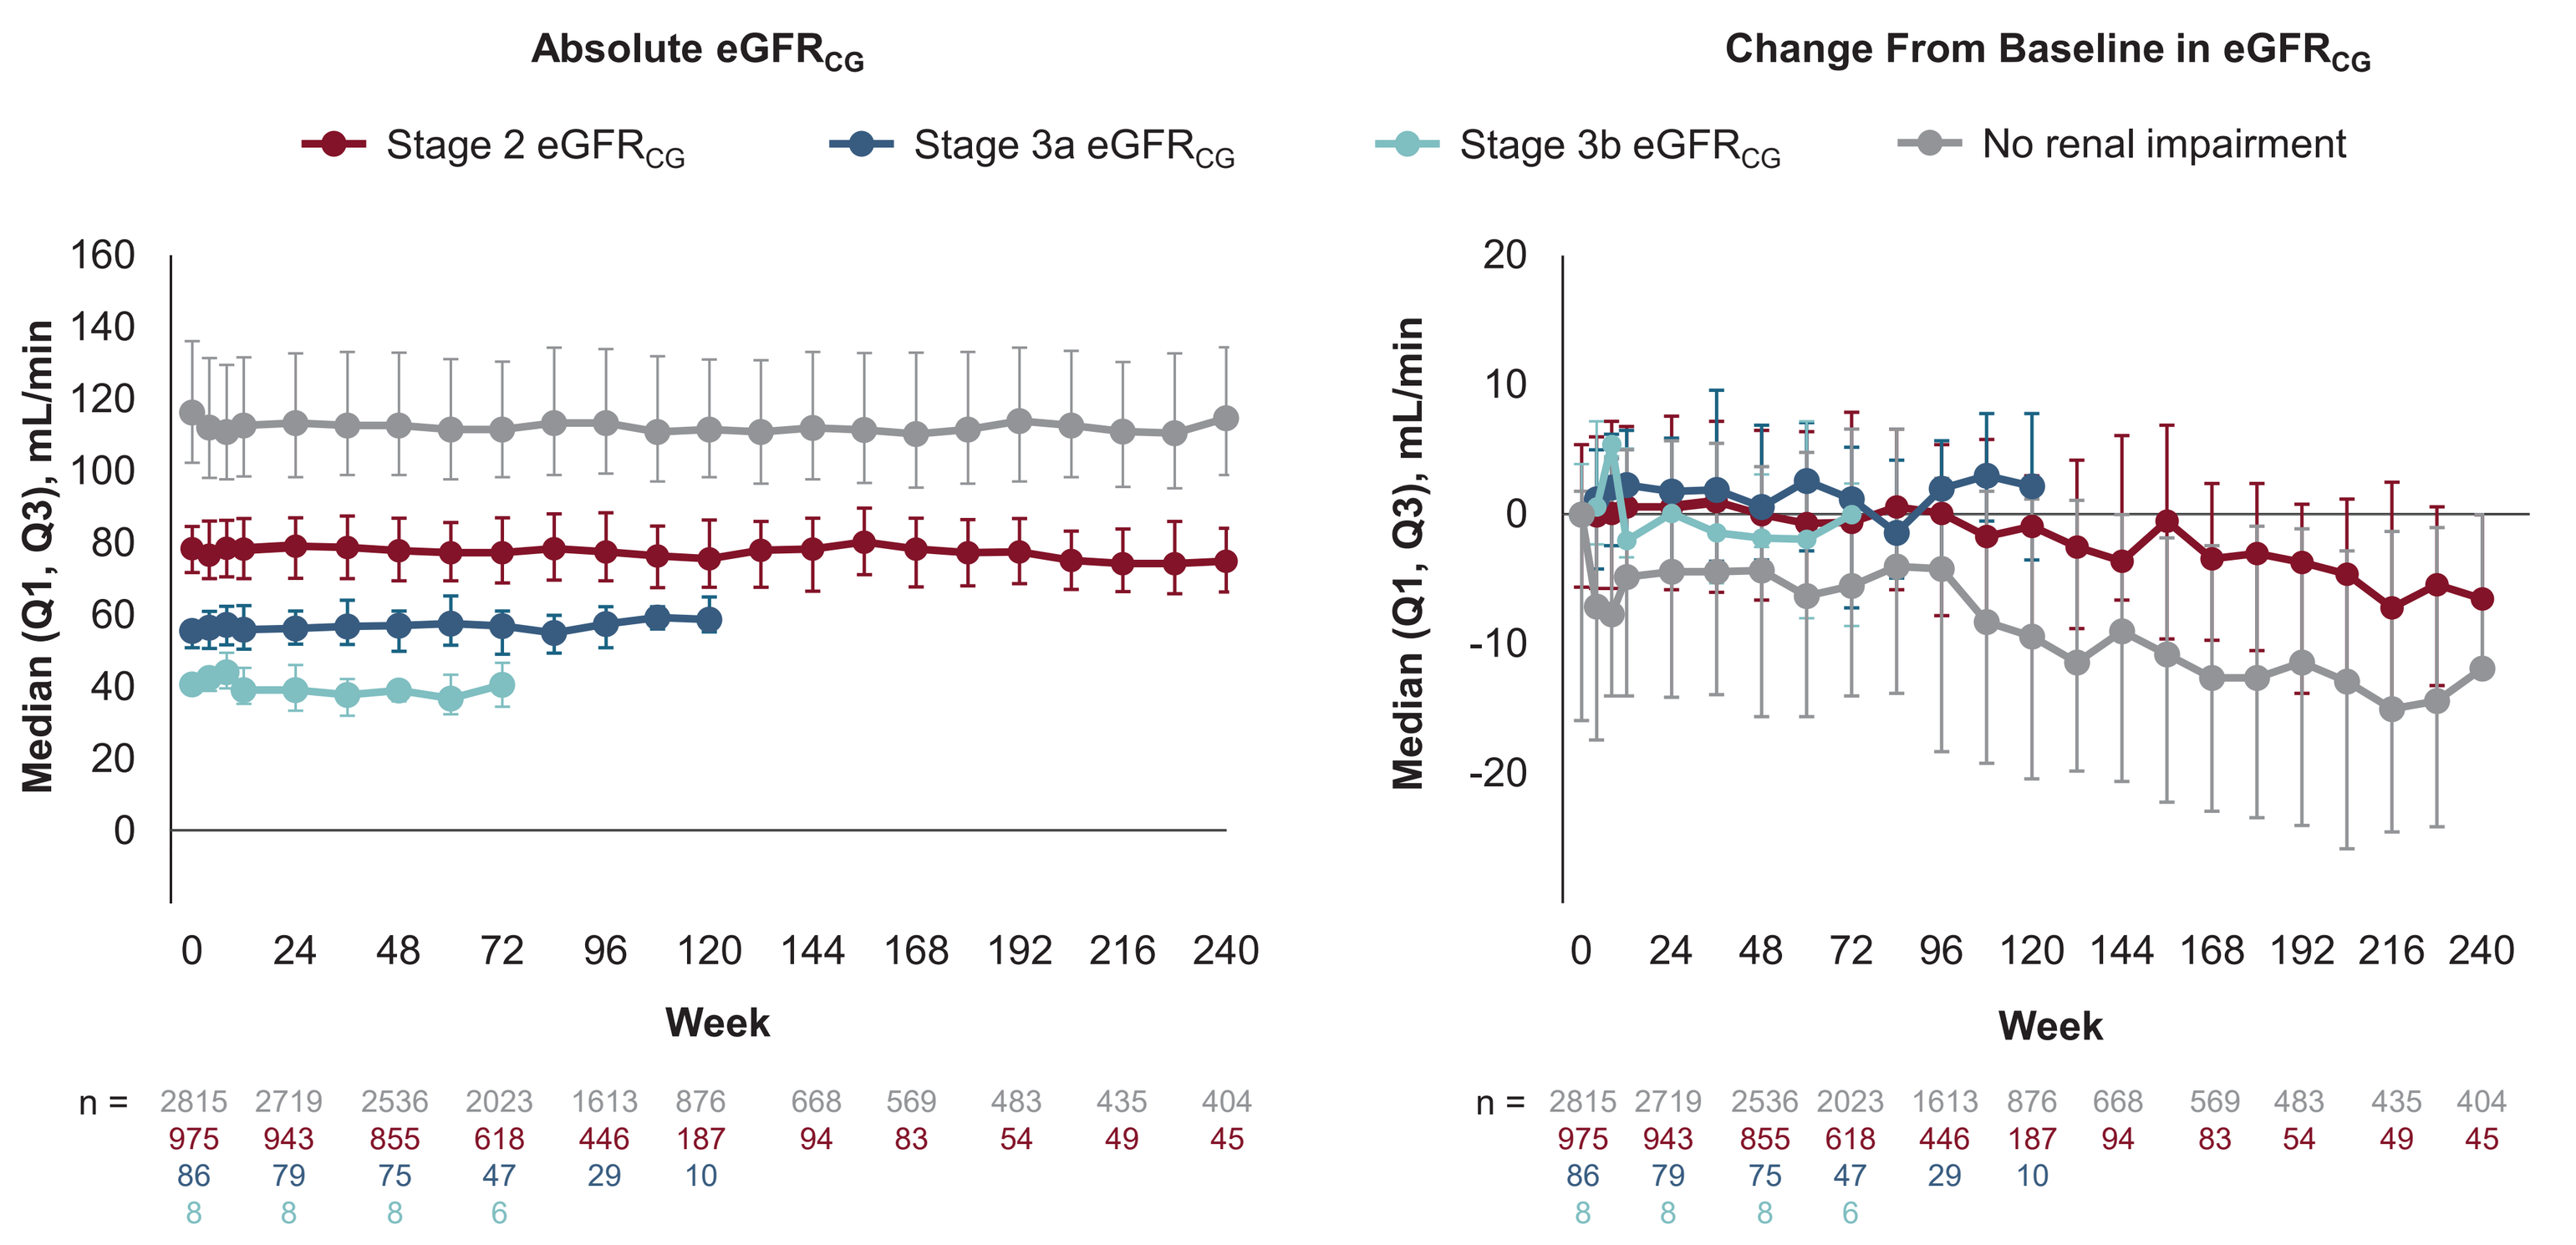
Absolute eGFRCG
Change From Baseline in eGFRCG
Stage 2 eGFRCG
Stage 3a eGFRCG
Stage 3b eGFRCG
No renal impairment
Median (Q1, Q3), mL/min
Week
0 24 48 72 96 120 144 168 192 216 240
n = 2815 2719 2536 2023 1613 876 668 569 483 435 404
975 943 855 618 446 187 94 83 54 49 45
86 79 75 47 29 10
8 8 8 6

Timepoints are shown for up to 5 years (240 weeks) or the last timepoint at which the number of participants with data was ≥ 5.
$eGFR_{CG}$: Stage 2, 60–< 90 mL/min; Stage 3a, 45–< 60 mL/min; Stage 3b, 30–< 45 mL/min; no renal impairment, ≥ 90 /mL/min.
$eGFR_{CG}$, estimated glomerular filtration rate by Cockcroft-Gault equation; Q, quartile.

Median Change From Baseline in $eGFR_{CG}$

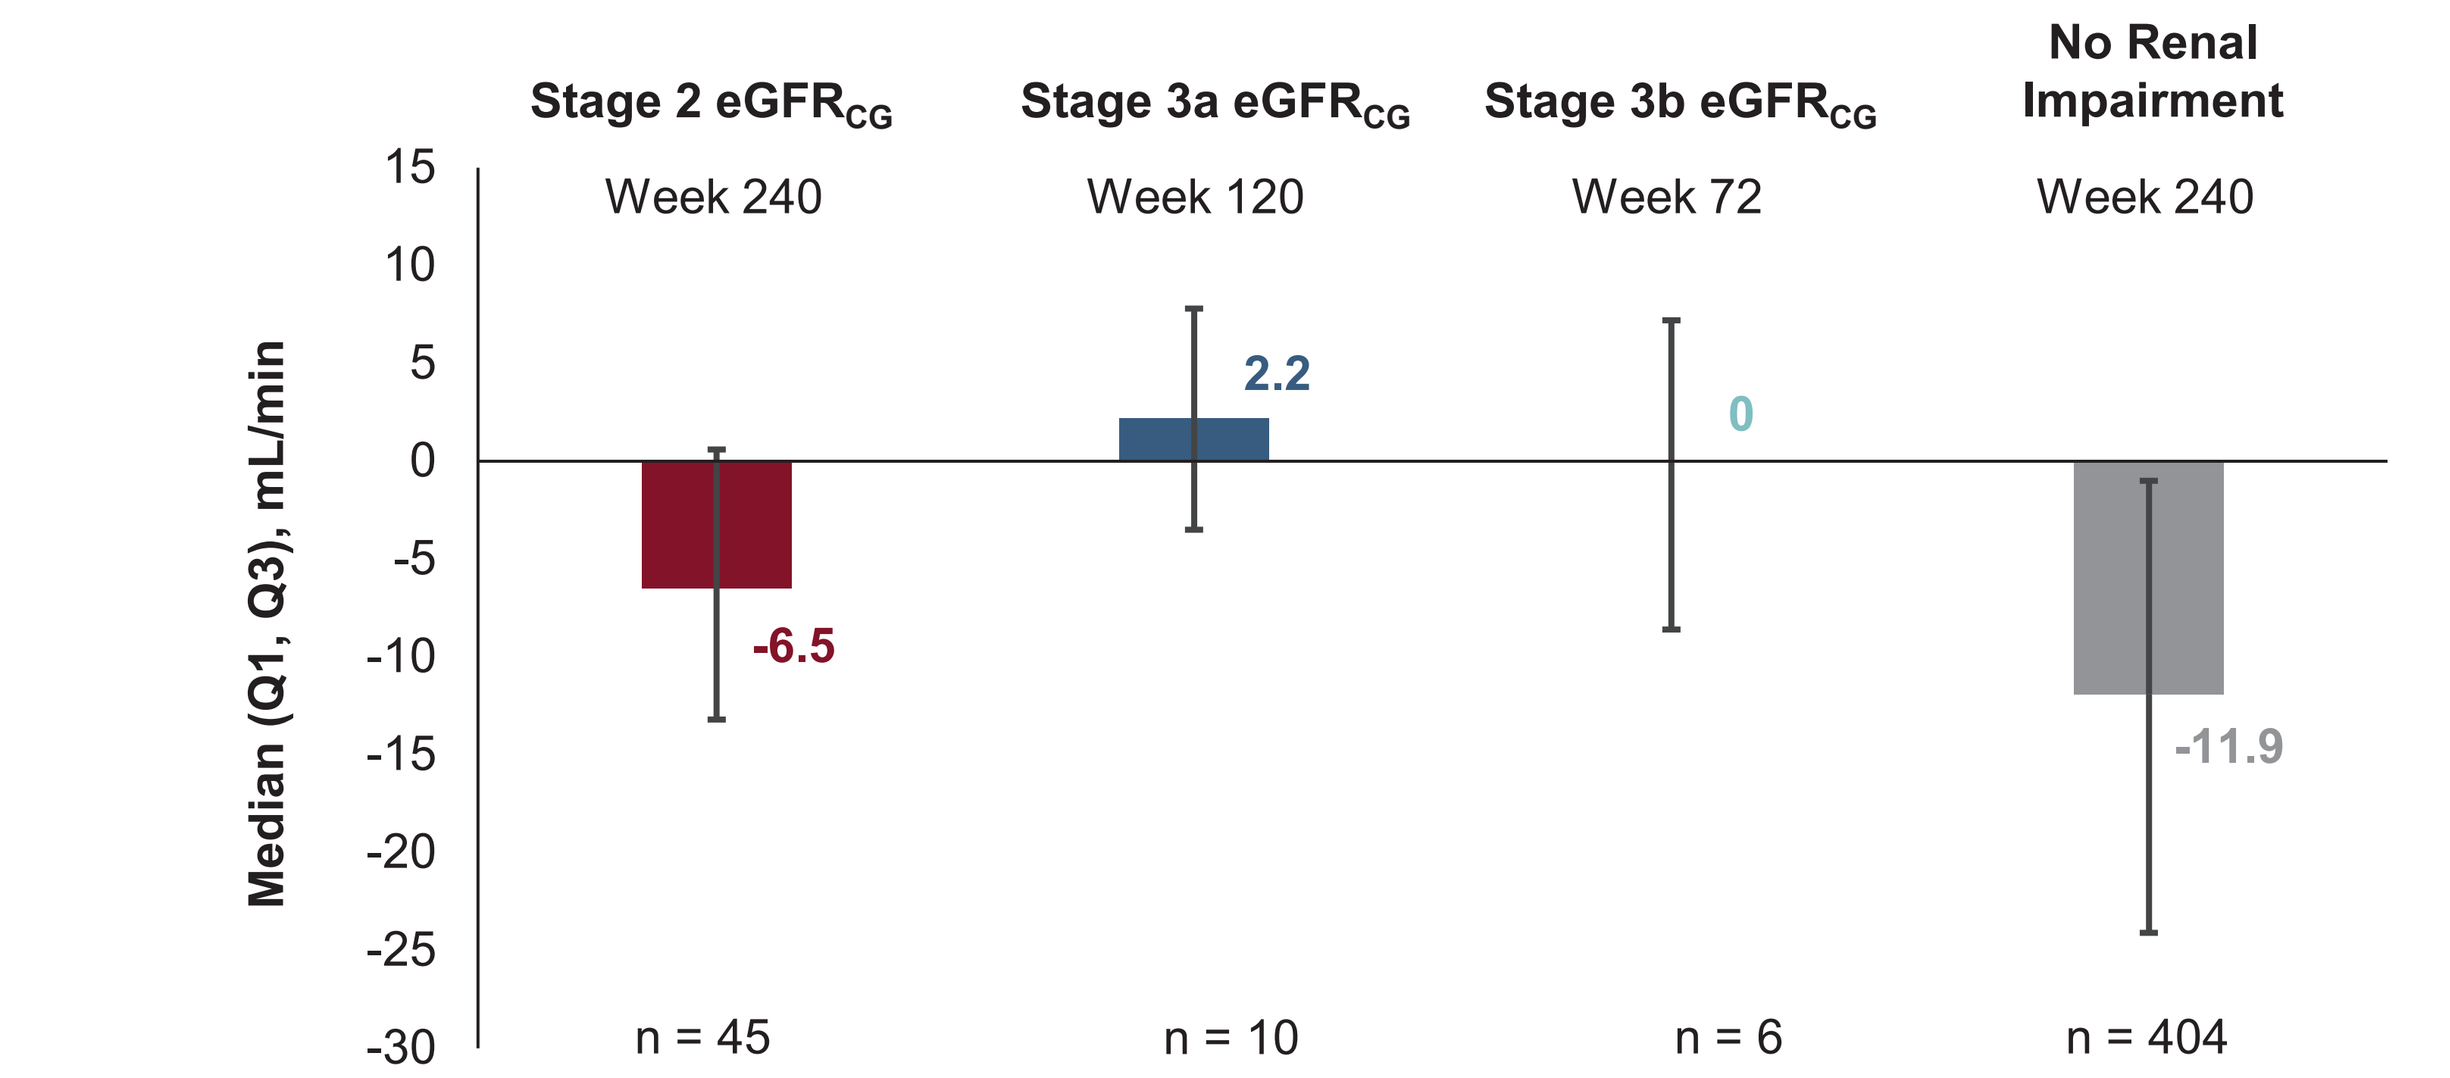
Stage 2 eGFRCG
Week 240
Stage 3a eGFRCG
Week 120
Stage 3b eGFRCG
Week 72
No Renal Impairment
Week 240
Median (Q1, Q3), mL/min
-6.5
2.2
0
-11.9
n = 45
n = 10
n = 6
n = 404

Timepoints are shown for up to 5 years (240 weeks) or the last timepoint at which the number of participants with data was ≥ 5.
$eGFR_{CG}$: Stage 2, 60–< 90 mL/min; Stage 3a, 45–< 60 mL/min; Stage 3b, 30–< 45 mL/min; no renal impairment, ≥ 90 /mL/min.
$eGFR_{CG}$, estimated glomerular filtration rate by Cockcroft-Gault equation; Q, quartile.

Change From Baseline in eGFR

- eGFR remained stable in all groups, with a small reduction in $eGFR_{CG}$ among participants with Stage 2 $eGFR_{CG}$ (median [Q1, Q3] change: -6.5 [-13.2, 0.6] mL/min at Week 240) and participants without renal impairment (median [Q1, Q3] change: -11.9 [-24.1, -1.0] mL/min at Week 240)

Renal TEAEs

	$eGFR_{CG}$ 30–< 90 mL/min (Renal Impairment) n = 1069	$eGFR_{CG}$ ≥ 90 mL/min (No Renal Impairment) n = 2815
Any renal TEAE, n (%)	28 (2.6)	71 (2.5)
Acute kidney injury	10 (0.9)	14 (0.5)
Creatinine renal clearance decreased	6 (0.6)	5 (0.2)
Proteinuria	5 (0.5)	39 (1.4)
Blood creatinine increased	4 (0.4)	8 (0.3)
Renal impairment	4 (0.4)	0
Renal failure	2 (0.2)	4 (0.1)
Glomerular filtration rate decreased	1 (< 0.1)	0
Protein urine present	0	4 (0.1)
Albuminuria	0	2 (< 0.1)
Blood creatinine abnormal	0	1 (< 0.1)
Blood urea increased	0	1 (< 0.1)
Urine output decreased	0	1 (< 0.1)

$eGFR_{CG}$, estimated glomerular filtration rate by Cockcroft-Gault equation; TEAE, treatment-emergent adverse event.

Renal TEAEs in Participants Receiving B/F/TAF

- Overall, 28 (2.6%) participants with renal impairment reported renal treatment-emergent adverse events (TEAEs); 7 (0.7%) were related to B/F/TAF
- Renal TEAEs occurred in 71 (2.5%) of participants without renal impairment, with 22 (0.8%) related to B/F/TAF
- The most common renal TEAEs were acute kidney injury (n = 10, 0.9%) in participants with renal impairment and proteinuria (n = 39, 1.4%) in participants without renal impairment
 - There were no cases of proximal renal tubulopathy or Fanconi syndrome

Discontinuation of B/F/TAF Due to Renal TEAEs

	$eGFR_{CG}$ 30–< 90 mL/min (Renal Impairment) n = 1069	$eGFR_{CG}$ ≥ 90 mL/min (No Renal Impairment) n = 2815
Renal TEAE leading to B/F/TAF discontinuation, n (%)	1 (< 0.1)	0
Acute kidney injury	1 (< 0.1)	0

B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; $eGFR_{CG}$, estimated glomerular filtration rate by Cockcroft-Gault equation; TEAE, treatment-emergent adverse event.

- **Only 1 (< 0.1%) participant discontinued B/F/TAF after 3 months due to a moderate renal TEAE of acute kidney injury, judged by investigator to be unrelated to B/F/TAF**
 - The participant was a 68-year-old male with Stage 2 $eGFR_{CG}$ and HIV-1 RNA < 50 copies/mL at baseline. They were receiving therapy for ongoing hyperlipidemia and hypertension at baseline, as well as an anticancer treatment with known association with kidney complications prior to discontinuation

Limitations

- Cystatin C–based eGFR was not evaluated, as in all trials included in the study, eGFR was calculated using the Cockcroft-Gault equation

References: **1.** Gallant J, et al. *Lancet*. 2017;390:2063-72. **2.** Sax PE, et al. *Lancet*. 2017;390:2073-82. **3.** Avihingsanon A, et al. *Lancet HIV*. 2023;10:e640-52. **4.** Molina J-M, et al. *Lancet HIV*. 2018;5:e357-65. **5.** Daar ES, et al. *Lancet HIV*. 2018;5:e347-56. **6.** Kityo C, et al. *J Acquir Immune Defic Syndr*. 2019;82:321-8. **7.** Sax PE, et al. *Clin Infect Dis*. 2021;73:e485-93. **8.** Maggiolo F, et al. *Infect Dis Ther*. 2021;10:775-88. **9.** Hagins D, et al. *J Acquir Immune Defic Syndr*. 2021;88:86-95. **10.** US Department of Health and Human Services. https://www.hiv.gov/hiv-basics/living-well-with-hiv/taking-care-of-yourself/aging-with-hiv (accessed Dec. 6, 2024). **11.** McCutcheon K, et al. *Circ Res*. 2024;134:1636-60. **12.** Surial B, et al. *J Infect Dis*. 2020;222:637-45. **13.** Gupta SK, et al. *AIDS*. 2019;33:1455-65. **14.** Biktarvy USPI, Gilead Sciences, Inc., April 2024.

Acknowledgments: We thank all study participants, investigators, and staff. This study was sponsored by Gilead Sciences, Inc. Medical writing support was provided by Flint Stevenson-Jones, PhD (Aspire Scientific Ltd, UK), and was funded by Gilead Sciences, Inc.

Disclosures: **FP** reports grants from Gilead Sciences, Inc., MSD, and ViiV Healthcare; payment or honoraria for lectures/education presentations from Gilead Sciences, Inc. and ViiV Healthcare; and conference attendance/travel reimbursements from MSD. **DW** reports grants or contracts from Gilead Sciences, Inc. and ViiV Healthcare; consulting fees from EMD Serono, Gilead Sciences, Inc., Merck, and ViiV Healthcare. **IB** reports grants or contracts and payment or honoraria from Gilead Sciences, Inc. and ViiV Healthcare. **AA** reports research/grant support from Gilead Sciences, Inc., GSK/ViiV Healthcare, Janssen, MSD, and Roche; travel support from Gilead Sciences, Inc.; and membership with the Committee of Thai AIDS Society, the Strategic and Technical Advisory Group on HIV, Hepatitis and STI for WHO SEARO, and the Thailand National Committee on ART, Hepatitis and TB/TPT.

Disclosures (continued): **HL**, **KA**, and **JTH** are employees of, and own stocks/shares in, Gilead Sciences, Inc. **SG** reports grants from ViiV Healthcare; and consulting fees from Gilead Sciences, Inc. and ViiV Healthcare. **GL**, **DH**, and **YY** have nothing to declare.

Correspondence: Frank Post, frank.post@kcl.ac.uk.

Conference on Retroviruses and Opportunistic Infections (CROI); March 9–12, 2025; San Francisco, CA, USA